II-01

POPULATION PHARMACOKINETIC MODELLING OF THE ANTIHISTAMINE BILASTINE IN CHILDREN WITHIN THE CONTEXT OF A MODEL INFORMED PAEDIATRIC DRUG DEVELOPMENT APPROACH

V. Vozmediano[1,2], E. Encinas[1], M. Rodriguez[1], JC. Lukas[1], N. Leal[1], M. Panselina[1,3], R. Valiente[4].

[1] Drug Modeling & Consulting, DynaKin S.L., Bilbao, Spain.
[2] Center for Pharmacometrics and Systems Pharmacology, University of Florida.
[3] Department of Pharmacology, University of the Basque Country, Spain.
[4] Clinical Research Department, FAES FARMA S.A., Leioa (Bizkaia), Spain.

BACKGROUND

Bilastine is a non-sedating second-generation H1-antihistamine approved for treatment of allergic rhinoconjunctivitis (AR) and urticaria (U) in adults at the oral (p.o.) dose of 20 mg once daily (OD) [1].

Optimal attributes can be anticipated for its clinical use in paediatrics due to its favourable safety and tolerability [2] and age-independent PD profile [3].

PURPOSE

The aim of this work was double:

1) To characterize bilastine PK in children through population modelling of data from clinical trial BILA-3009/PED ("Multicenter, International, Adaptive, Open-label, Repeated Administration PK Study of bilastine (10 mg/day) in Children from 2 to <12 years with AR or U") as part of bilastine paediatric drug development.

2) To ascertain whether the proposed dose in the above paediatric subset (10 mg/day) matches systemic exposure seen in adults at the 20 mg/day dose.

METHODS

BILA-3009/PED was an adaptive limited sampling confirmatory study, optimized by means of an ontogenic predictive PK/PD model developed in children between 2 and 12 years based on prior knowledge [3].

The paediatric PK clinical dataset comprised a total of 31 patients from 4 to <12 years treated with 10 mg oral bilastine daily. Sparse PK sampling (i.e., maximum of 6 or 4 samples per child, in ≥6 and <6 year old patients, respectively) was performed up to 24 h following administration of the 6th dose.

Population PK (PopPK) models were developed in NONMEM (FOCE method). Graphical analysis was performed in S-PLUS.

One and two compartment disposition models with first-order absorption and possible contribution of a lag time were explored.

The influence of covariates (e.g. age, body weight (WT), height, sex and creatinine clearance (CrCl)) was then evaluated by a stepwise procedure.

Final model establishment and validation was done using standard statistical and diagnostic criteria for parametric non-linear mixed effects models, including goodness of fit (GOF) plots, visual predictive check (VPC) and posterior predictive check (PPC).

Suitability of the final paediatric dataset to fulfil the trial stopping criteria (i.e., model completeness and non-dependence of exposure on decreasing age) was also evaluated.

In a final step, the popPK model along with observations and additional analyses (including e.g. a physiological interpretation of bilastine PK parameters and allometric scaling methods) were used to assess the suitability of the selected paediatric dose (10 mg/day) through comparison of PK metrics in children with those in adults (20 mg/day).

RESULTS

A two-compartment disposition model (same structure as for bilastine PK in adults [4]) with a lag time and proportional residual error provided the best description of the paediatric observations. The interindividual variability (modelled as exponential) was well defined and significant for Ka and CL/F. Population parameter estimates together with the standard error of estimates (SEE) are presented in *Table 1*. The achievement of convergence in FOCE method along with adequate SEE in the model parameters, are indicators of the suitability of the data to provide sufficient model identification.

None of the studied covariates was significantly predictive for inclusion [*Table 2*].

Table 1. Bilastine population PK final model parameters in children.

Parameter	Estimate	SEE (%)
Ka (h-1)	1.29	22.2
CL/F (L/h)	12.5	5.9
Vc/F (L)	19.7	12.8
Q/F (L/h)	2.01	34
Vp/F (L)	17.4	14.8
Tlag (h)	0.183	4.3
ωCL (%)	15	40
ωKa (%)	67	30
σ (%)	35	22

Ka: first-order absorption rate constant, CL/F: apparent clearance, Vc: central volume of distribution, Q: intercompartmental clearance, Vp: peripheral volume of distribution, Tlag: lag time, ω: interindividual variability, σ: residual variabiliy.

Table 2. Covariate models tested during data analysis of trial BILA-3009/PED.

Covariate added	Equation used for covariate inclusion	OBJ	Delta OBJ	Comment
None – base model	NA	597.751	NA	Base model
WT on CL	$CL_{po} = \theta_{CLtv} \cdot \left(\frac{Weight}{70}\right)^{0.63}$	608.961	11.21	Not significant
WT on CL	$CL_{po} = \theta_{CLtv} \cdot \left(\frac{Weight}{70}\right)^{0.75}$	613.397	15.646	Not significant
WT on CL	$CL_{po} = \theta_{CLtv} \cdot \left(\frac{Weight}{70}\right)^{PWR1}$	597.548	-0.203	Not significant Allometric exponent 0.0715 (SEE 178%)
Standardized CrCl and WT on CL	$CL_{po} = \theta_{CLtv} \cdot CLCr_{std}$ $CLCr_{std} = (CLCr \cdot \left(\frac{70}{WT}\right))/100$ Note: 100 mL/min 'normal' GFR for 70kg	612.419	14.668	Not significant
CrCl and WT on CL	$CL_{po} = \left(\theta_{cl1} + \theta_{cl2} \cdot \left(\frac{CLCr}{100}\right)\right) \cdot \left(\frac{Weight}{70}\right)^{0.75}$	613.407	15.656	No SEE Not significant
WT and age on CL	$CL_{po} = \theta_{CLtv} \cdot \left(\frac{Weight}{70}\right)^{PWR1} \cdot \left(\frac{Age}{24}\right)^{PWR2}$	596.588	-1.163	No SEE Not significant
WT and age on CL	$CL_{po} = \theta_{CLtv} \cdot \left(\frac{Weight}{70}\right)^{0.75} \cdot \left(\frac{Age}{24}\right)^{PWR1}$	613.676	15.925	No SEE Not significant
Age on CL	$CL_{po} = \theta_{CLtv} \cdot \left(\frac{Age}{24}\right)^{PWR1}$	596.534	-1.217	Not significant Power exponent 0.133 (SEE 71.70%)
Mean standard children age on CL	$CL_{po} = \theta_{CLtv} \cdot \left(\frac{Age}{6}\right)^{PWR1}$	596.531	-1.217	Not significant Power exponent 0.133 (SEE 69.4%)
WT and age on CL	$CL_{po} = \theta_{CLtv} \left(\frac{Weight}{70}\right)^{PWR1} \cdot \left(1 - \frac{Age}{24}\right)^{PWR2}$	597.548	-0.203	Not significant Allometric exponent 0.0715 (SEE 201%) Power exponent on age 22.9 (SEE 1389%)
WT on CL	$CL_{po} = \theta_{CL1} + \theta_{CL2} \cdot \left(\frac{Weight}{70}\right)$	597.292	-0.459	Not significant

Final popPK model was positively qualified by diagnostic GOF plots and its predictive capacity was confirmed at all ages recruited via VPC [*Figure 1*] and PPC [*Figure 2*]. The model was proven complete and independent of age and WT, supporting that the entire paediatric group [4 to <12 years] belongs to the same PK population. Stopping rules pre-stablished to finalize the trial were thus fulfilled.

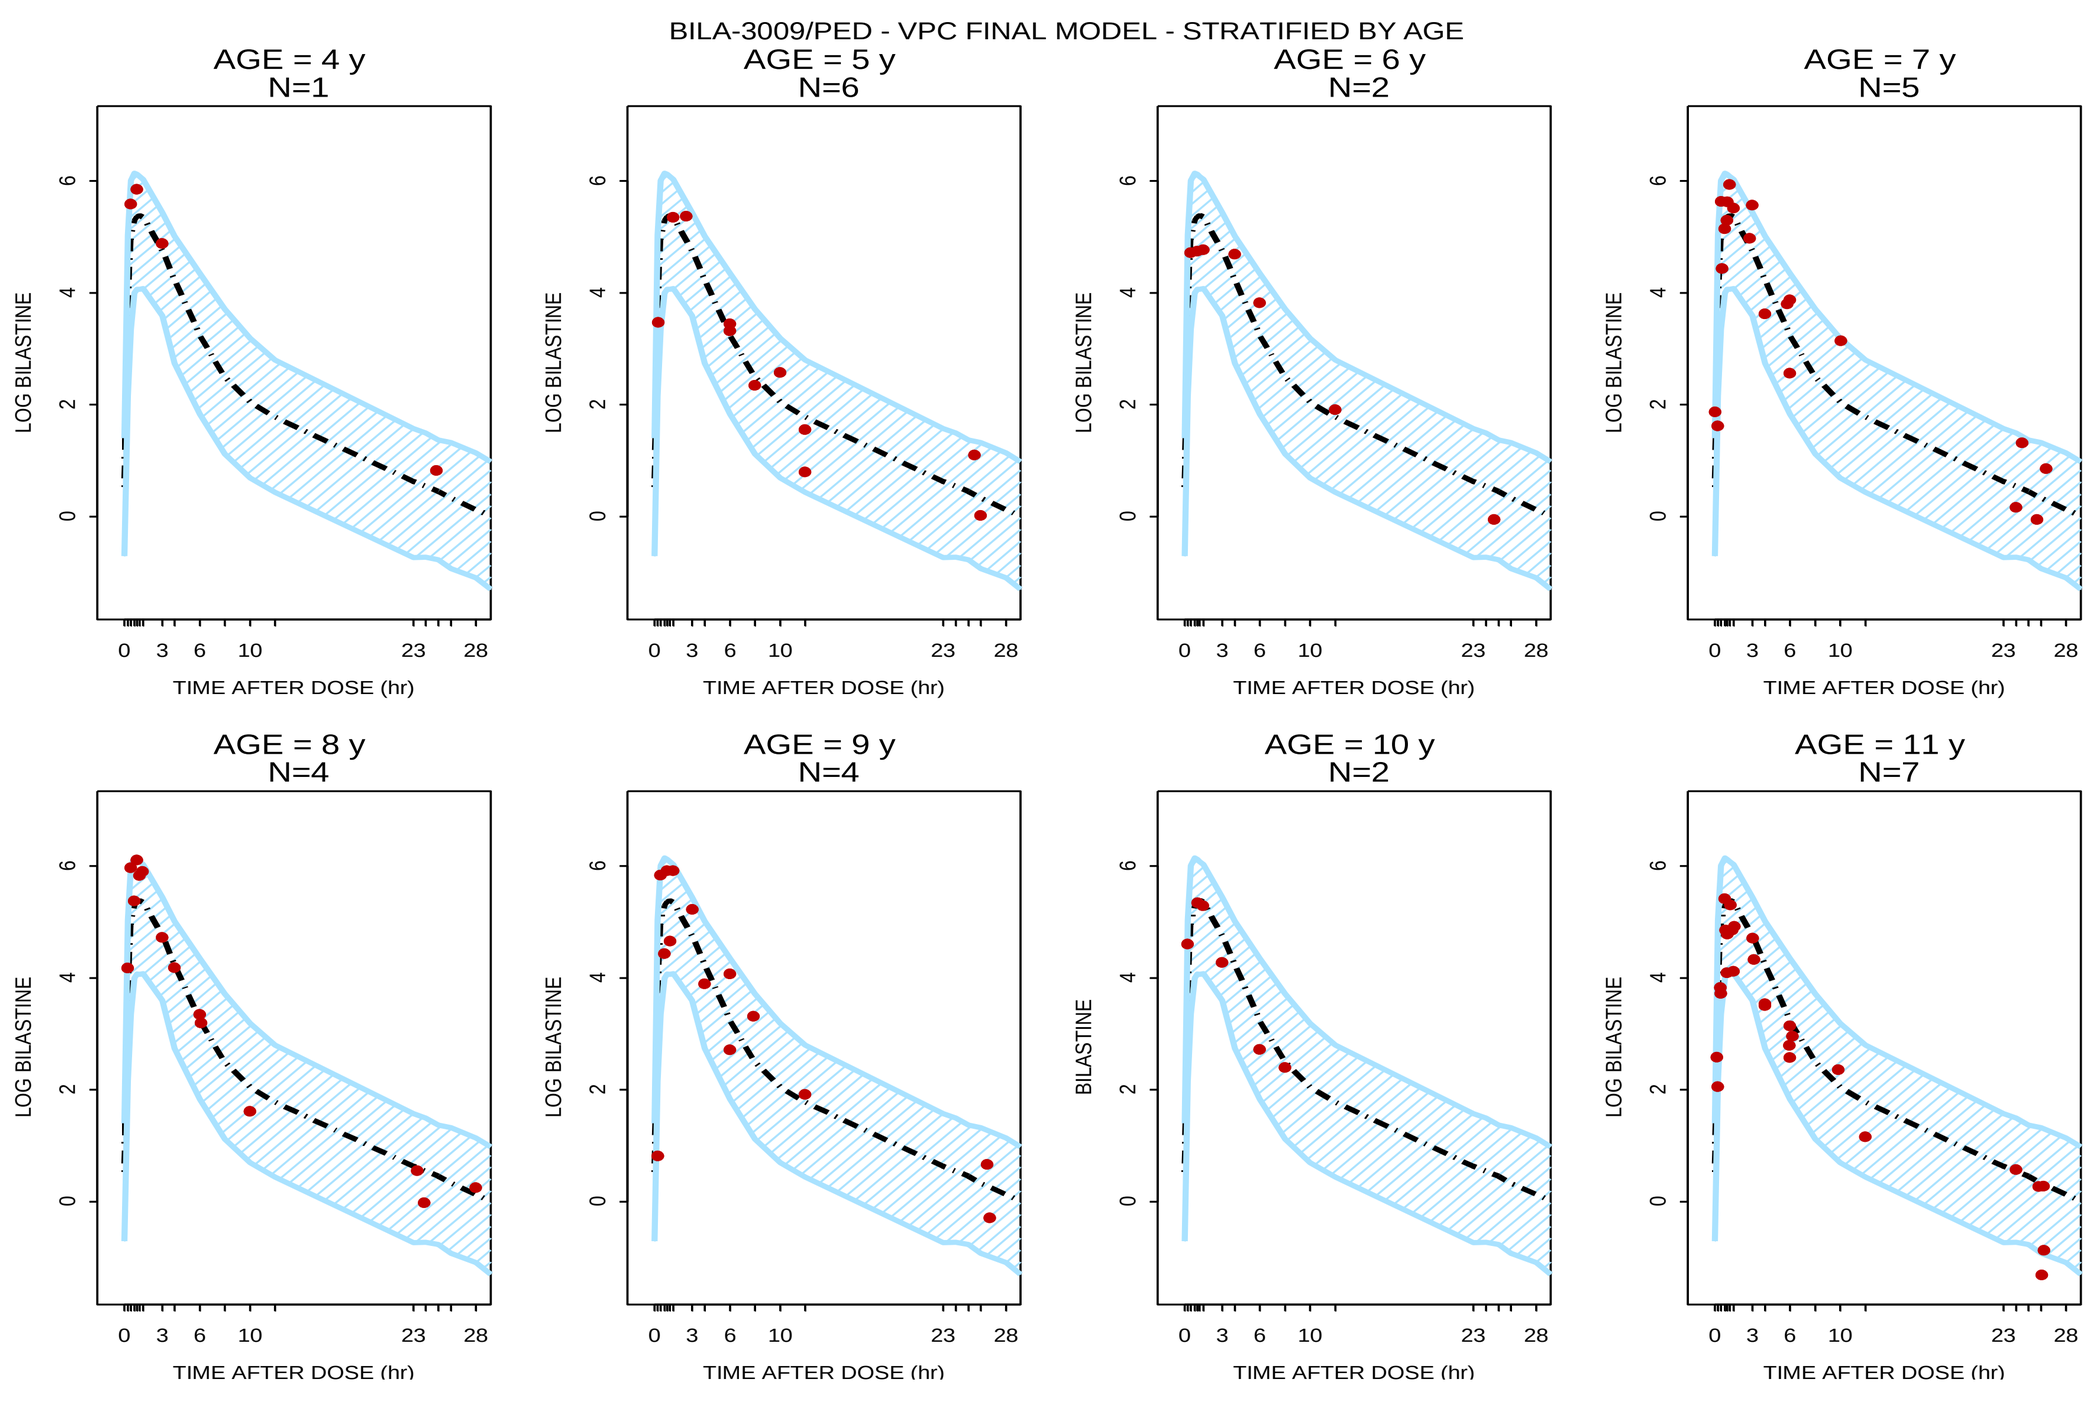
BILA-3009/PED - VPC FINAL MODEL - STRATIFIED BY AGE
AGE = 4 y
N=1
AGE = 5 y
N=6
AGE = 6 y
N=2
AGE = 7 y
N=5
AGE = 8 y
N=4
AGE = 9 y
N=4
AGE = 10 y
N=2
AGE = 11 y
N=7
LOG BILASTINE
TIME AFTER DOSE (hr)

Figure 1. Evaluation of the final pediatric popPK model via VPC stratified by age. Plasma concentrations are in Log scale. Observation (red dots) are overlaid on the PK predictive intervals (mean, 95% CI) simulated by the model after the steady state administration of 10 mg of bilastine in children (N=1000).

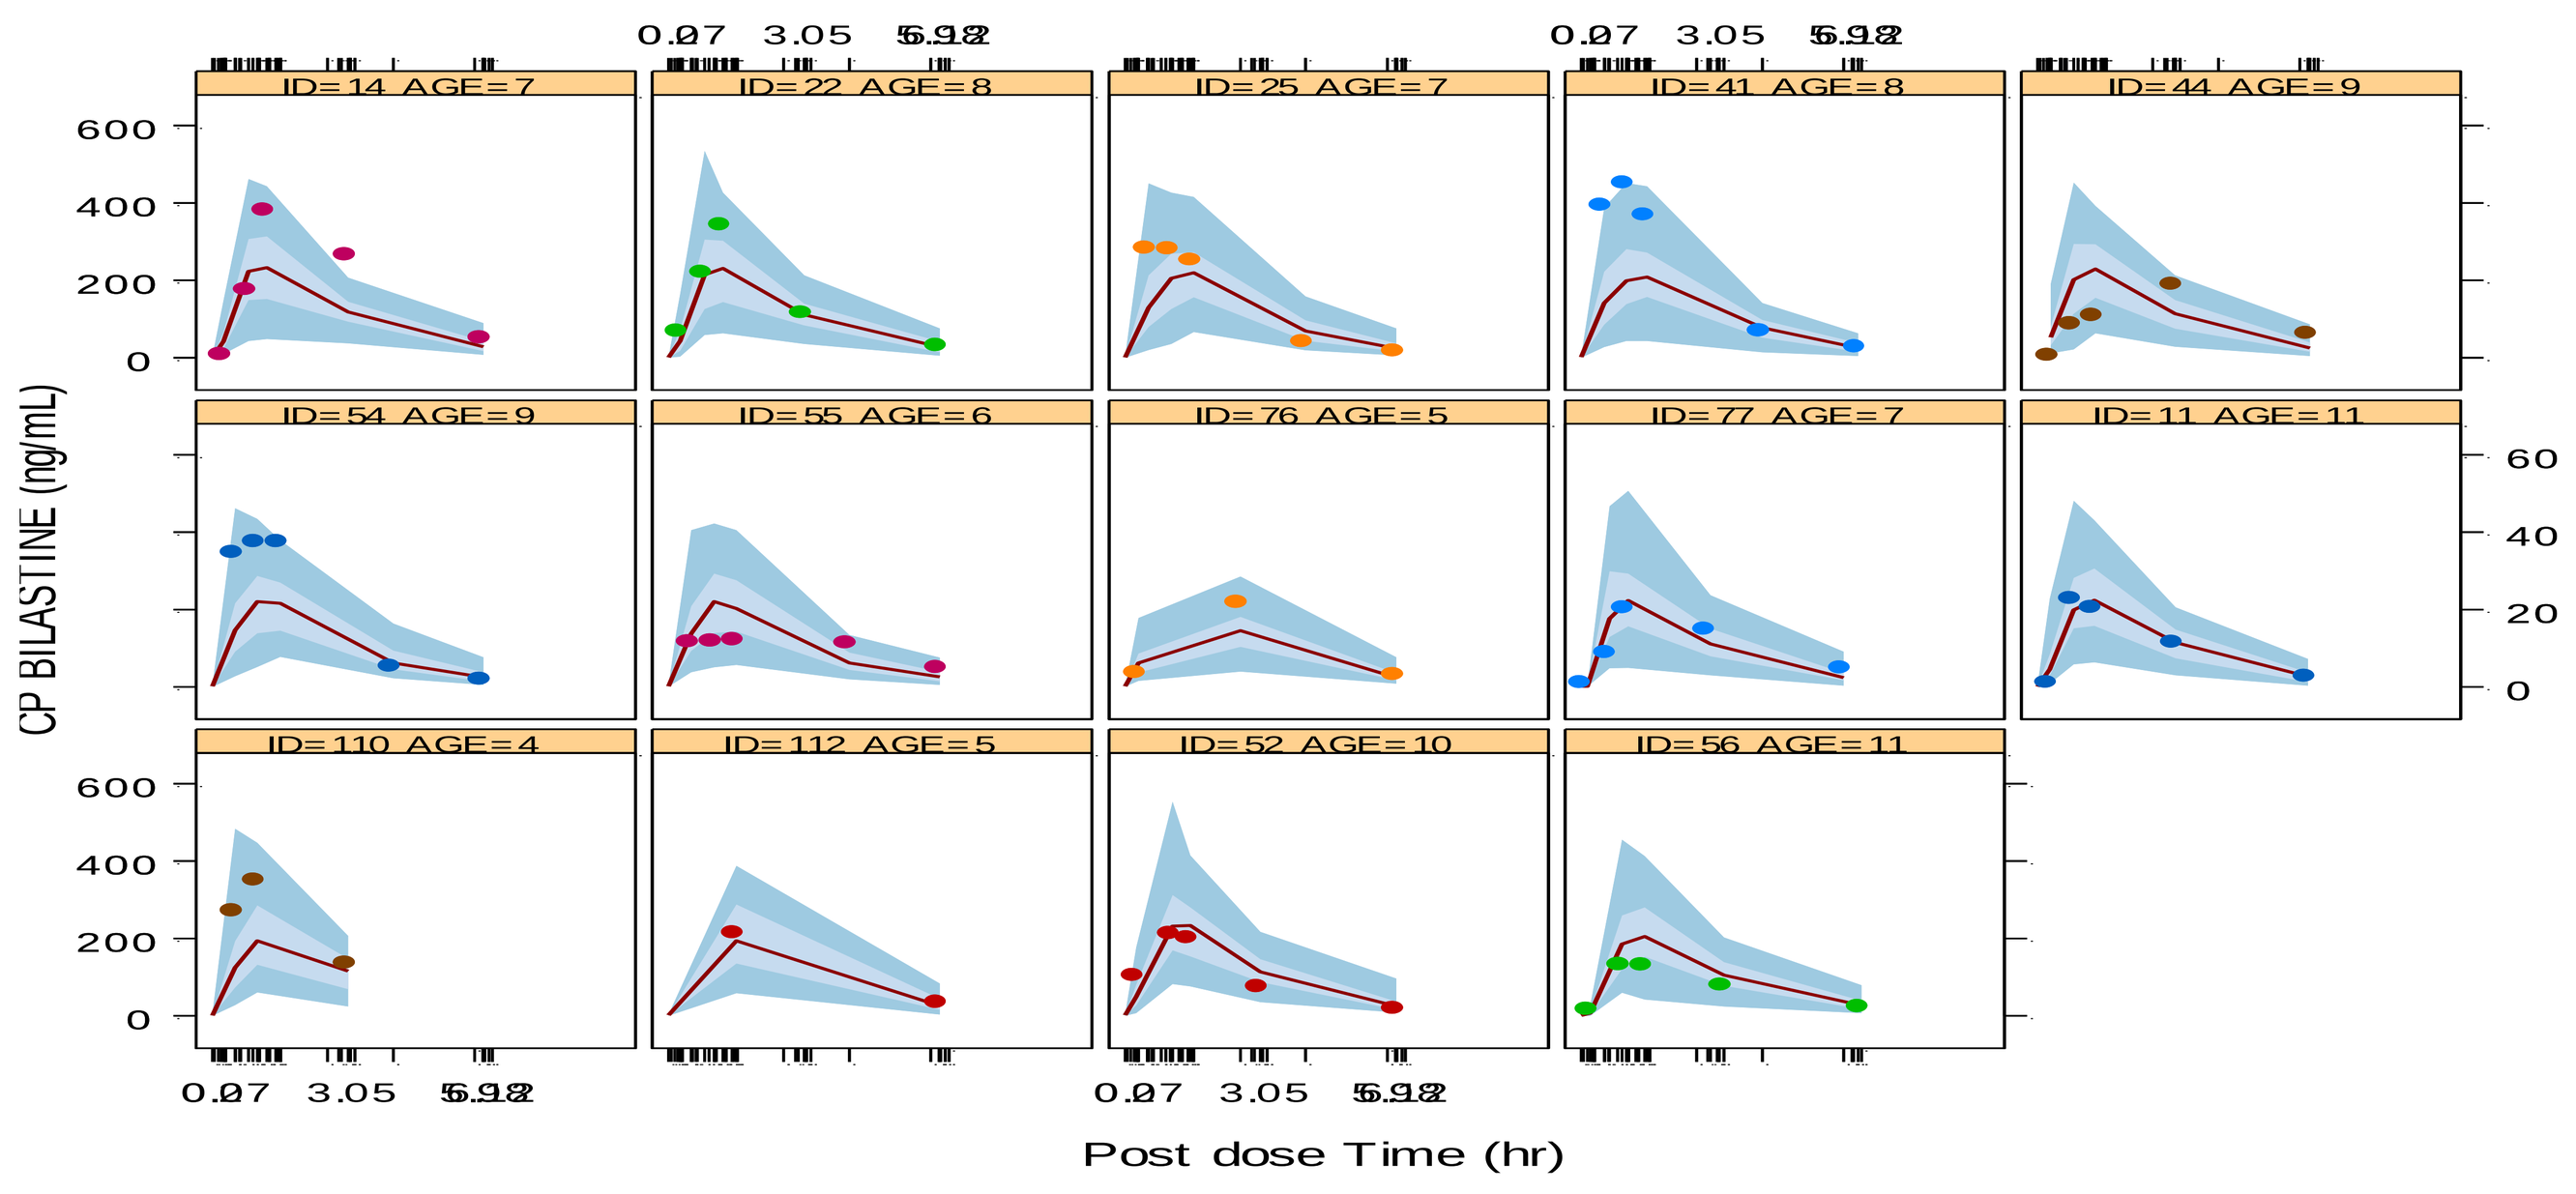
ID=14 AGE=7
ID=22 AGE=8
ID=25 AGE=7
ID=41 AGE=8
ID=44 AGE=9
ID=54 AGE=9
ID=55 AGE=6
ID=76 AGE=5
ID=77 AGE=7
ID=11 AGE=11
ID=110 AGE=4
ID=112 AGE=5
ID=52 AGE=10
ID=56 AGE=11
CP BILASTINE (ng/mL)
Post dose Time (hr)

Figure 2. Evaluation of the final pediatric popPK model via PPC. Posterior population simulations from the parameter uncertainty and the random variable distributions are shown, including mean prediction (solid red line), 50% CI and 95% CI (light and dark shades). Observations are overlaid colour coded arbitrarily by subject for N=14 IDs with non-trivial profiles (more than single time-point sampled).

Children from 2 years were also deemed to belong to the same PK population as well, given that the physiology concerned is already mature by this age, as further supported by a number of additional analyses (including confirmation of ontogenic assumptions and physiological interpretation of bilastine PK processes) [*Table 3*].

Table 3. Summary of the predictable bilastine PK behaviour in children >2 years from the perspective of a physiological and ontogenic interpretation.

	Well Defined in Adult	Well Defined in Children	Ontogenic Differences
PK linear	✓	✓	Linearity demonstrated in adults in the range 2.5-220 mg. No unexpected non-linearities in children > 2 years with the 10 mg dose (age dependency as a function of body composition, organ functions and ontogeny of drug pathways relevant only during first 2 years of life).
Drug does not accumulate	✓	✓	Volume of distribution of bilastine is mainly related with total body water. TBW is ~61.2% of the body weight in children 1-2 years vs. 60% in adults. Therefore, no accumulation is expected. Differences in the body composition between children and adults were considered in the ontogenic model used to select the paediatric dose.
Rapid first order absorption	✓	✓	Bioequivalence between children and adults formulations demonstrated. No differences expected in absorption as the physiological functions of the gastrointestinal tract affecting bilastine (including levels and activity of P-glycoprotein and OATP active transporters) are mature by the age of 2.
Food Effect*	✓	✓	
Protein binding	✓	✓	The slight changes in the protein binding of children >2 years with respect to adult are not significant. Differences in protein binding were considered in the ontogenic approach.
Drug is not metabolized	✓	✓	Not applicable
Excreted by glomerular filtration	✓	✓	Renal function is already mature by the age of 2 years, and the capacity of glomerular filtration is similar in adults and children after this age.
Renal elimination proportional to the GFR	✓	✓	Renal clearance is proportional to the patient's renal function which is already mature by the age of 2 years. Bilastine renal clearance in children is considered proportional to individual glomerular filtration rate (GFR) values and the unbound fraction of the drug.

*Similar, independent on formulation

No significant difference was observed in AUC and C_{max} between paediatric age groups, all of which were well within the range observed in adults at the therapeutic dose (20 mg/day) [*Table 4*].

Table 4. Summary statistics of AUC and C_{max} observed in children from study BILA-3009/PED (at bilastine 10 mg/day) as compared to adults (at 20 mg/day).

	$AUC_{0\text{-}24h}$ (ng·h/mL)			C_{max} (ng/mL)		
Population	**Mean**	**Median**	**Min-Max**	**Mean**	**Median**	**Min-Max**
Children (<6 years)	1363	1022	958-2110	257	214	210-347
Children (≥6 years)	1073	1086	532-1653	255	224	118-447
Adults	1160	1121	481-2528	260	232	83-924

Moreover, the extrapolated exposures in 4 and 2 years old children using alternative scaling methods, i.e. a GFR-based approach as well as allometry, were well within the range of AUC observed in adults, and far from the safety threshold [*Table 5*].

Table 5. AUC (ng·h/mL) predicted in children of 2 and 4 years of age receiving 10 mg bilastine by using different extrapolation methods.

Scaling method	4 years	2 years
Allometry from adult - fixed exponent 0.63[1]	1603	1957
From the glomerular filtration rate (GFR)[2]	1154	1450

(1) Using the 5th percentiles of weights for 4 and 2 years olds (12.9 kg and 9.40 kg, respectively) [6]
(2) Using the ontogenic model [3]
Safety threshold: 4799 ng·h/mL [3]

All the above approaches supports the choice of 10 mg OD p.o. for the entire children subset, as further confirmed in a placebo-controlled safety trial including 509 children from 2 to <12 years (BILA-3312/PED) [5].

CONCLUSION

A popPK model characterizing bilastine PK behaviour in children aged from 4 to <12 years was successfully developed and positively qualified from observations in a limited sampling confirmatory study in paediatrics. Additionally, the achievement of comparable (i.e., within the range) drug exposure to that observed in adults after the therapeutic dose of 20 mg, together with the additional integrative analysis, served to confirm the validity of the 10 mg daily dose for the entire target paediatric subset (2 to <12 years).

REFERENCES

[1] Bachert C, Kuna P, Zuberbier T. Bilastine in allergic rhinoconjunctivitis and urticaria. Allergy. 2010;65:1–13.
[2] Sadaba B, Azanza JR, Gomez-Guiu A, Rodil R. Critical appraisal of bilastine for the treatment of allergic rhinoconjunctivitis and urticaria. Ther. Clin. Risk Manag. 2013;9:197–205.
[3] Vozmediano V, Sologuren A, Lukas JC, Leal N, Rodriguez M. Model Informed Pediatric Development Applied to Bilastine: Ontogenic PK Model Development, Dose Selection for First Time in Children and PK Study Design. Pharm Res. 2017 Dec;34(12):2720-2734.
[4] Jauregizar N, de la Fuente L, Lucero ML, Sologuren A, Leal N, Rodríguez M. Pharmacokinetic-pharmacodynamic modelling of the antihistaminic (H1) effect of bilastine. Clin. Pharmacokinet. 2009;48:543–54.
[5] Novák Z, Yáñez A, Ildikó K, Kuna P, Tortajada-Girbés M, Valiente R. Safety and tolerability of bilastine 10 mg administered for 12 weeks in children with allergic diseases. Pediatr Allergy Immunol. 2016 Aug;27(5):493-8.
[6] World Health Organization. WHO Child Growth Standards. 2006; Available from: http://www.who.int/childgrowth/standards/Technical_report.pdf